Phase I Study of Belantamab Mafodotin in Combination with Standard of Care in Transplant-Ineligible Newly Diagnosed Multiple Myeloma: DREAMM-9 Updated Interim Analysis

The 11th World Congress on CONTROVERSIES IN MULTIPLE MYELOMA (COMy)

Saad Z. Usmani, MD[1], Michał Mielnik, MD, PhD[2], Mamta Garg, MD, FRCP, FRCPath[3], Irwindeep Sandhu, MD[4], Al-Ola Abdallah, MD, PhD[5], Youngil Koh, MD[6], Albert Oriol, MD[7,8], Hang Quach, MD[9], Katja Weisel, MD, PhD[10], Aránzazu Alonso Alonso, MD[8,11], Enrique M. Ocio, MD, PhD[8], Wojciech Janowski, MD[12], Chang-Ki Min, MD[13], Karthik Ramasamy, MD[14], Ricarda Garcia Sanchez, MD[15], Paula Rodriguez-Otero, MD[16], Chris Brawley, MSc[17], Jacqueline L. Egger, PhD[17], Morrys C. Kaisermann, MD, PhD[18], and Marek Hus, MD, PhD[2]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Department of Hematooncology and Bone Marrow Transplantation, Medical University of Lublin, Poland; [3]Leicester Royal Infirmary, Leicester, UK; [4]University of Alberta, Edmonton, AB, Canada; [5]University of Kansas Medical Center, US Myeloma Research Innovations Research Collaborative (USMIRC), Westwood, KS, USA; [6]Department of Internal Medicine, Seoul National University Hospital, Seoul, South Korea; [7]Institut Català d'Oncologia and Institut Josep Carreras - Hospital Universitari Germans Trias i Pujol (HUGTP), Badalona, Spain; [8]Hospital Universitario Marqués de Valdecilla (IDIVAL), Universidad de Cantabria, Santander, Spain; [9]St Vincent's Hospital Melbourne, University of Melbourne, Melbourne, VIC, Australia; [10]University Medical Center of Hamburg-Eppendorf, Hamburg, Germany; [11]Hospital Quirón Madrid, Madrid, Spain; [12]Calvary Mater Newcastle, Newcastle, Australia; [13]Seoul St. Mary's Hospital, Seoul, South Korea; [14]Oxford Translational Myeloma Centre, NDORMS, University of Oxford and Churchill Hospital, Headington, Oxford, UK; [15]Department of Hematology, Hospital Universitario Virgen de la Victoria, Málaga, Spain; [16]Cancer Center Clínica Universidad de Navarra, Pamplona, Spain; [17]GSK, London, UK; [18]GSK, Upper Providence, PA, USA

INTRODUCTION

- Belantamab mafodotin (belamaf) is an ADC consisting of an afucosylated anti-BCMA monoclonal antibody and a cytotoxic payload, mcMMAF[1,2]
- The Phase 3 studies DREAMM-7 (BVd vs DVd) and DREAMM-8 (BPd vs PVd) showed significant improvement in PFS and manageable safety profiles in patients with RRMM[3,4]
- VRd is one of the SOC treatments for patients with NDMM without intent for SCT;[5,6] however, there remains a need for deeper and longer responses for patients with TI NDMM
- The DREAMM-9 study is evaluating multiple doses and schedules of belamaf + VRd in patients with TI NDMM[7,8]

AIMS

- Evaluate the safety and efficacy of belamaf + VRd in the TI NDMM setting

Figure 1: Belamaf multimodal mechanism of action

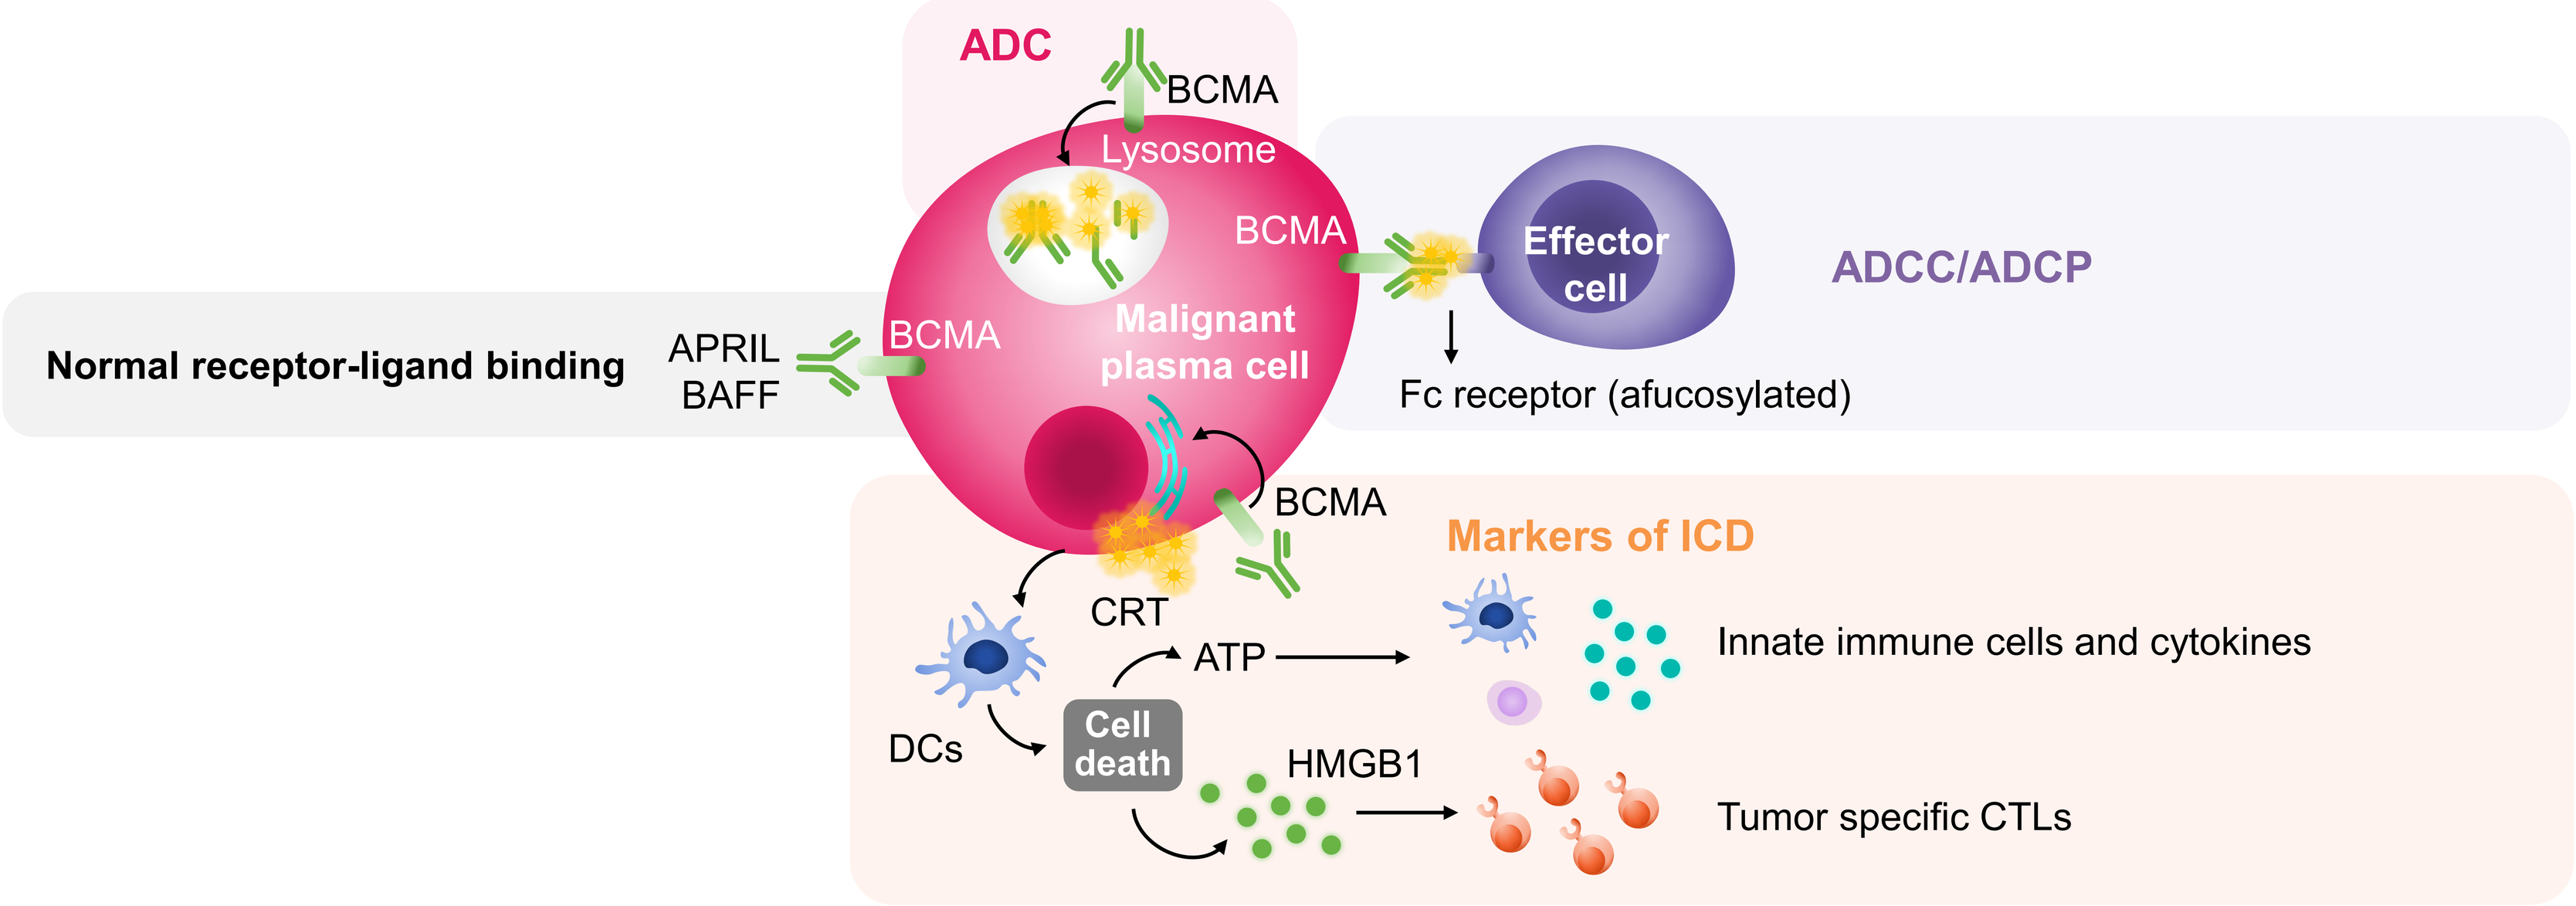
ADC
BCMA
Lysosome
BCMA
Effector cell
ADCC/ADCP
Normal receptor-ligand binding
APRIL
BAFF
BCMA
Malignant plasma cell
Fc receptor (afucosylated)
BCMA
Markers of ICD
CRT
ATP
Innate immune cells and cytokines
DCs
Cell death
HMGB1
Tumor specific CTLs

STUDY DESIGN

Inclusion criteria

- MM diagnosis per IMWG criteria
- ECOG performance status of 0–2
- Not a candidate for HDT with ASCT due to frailty and/or significant comorbid condition(s)
- Measurable disease per IMWG criteria

Exclusion criteria

- Prior systemic therapy for MM or SMM
- Current corneal epithelial disease

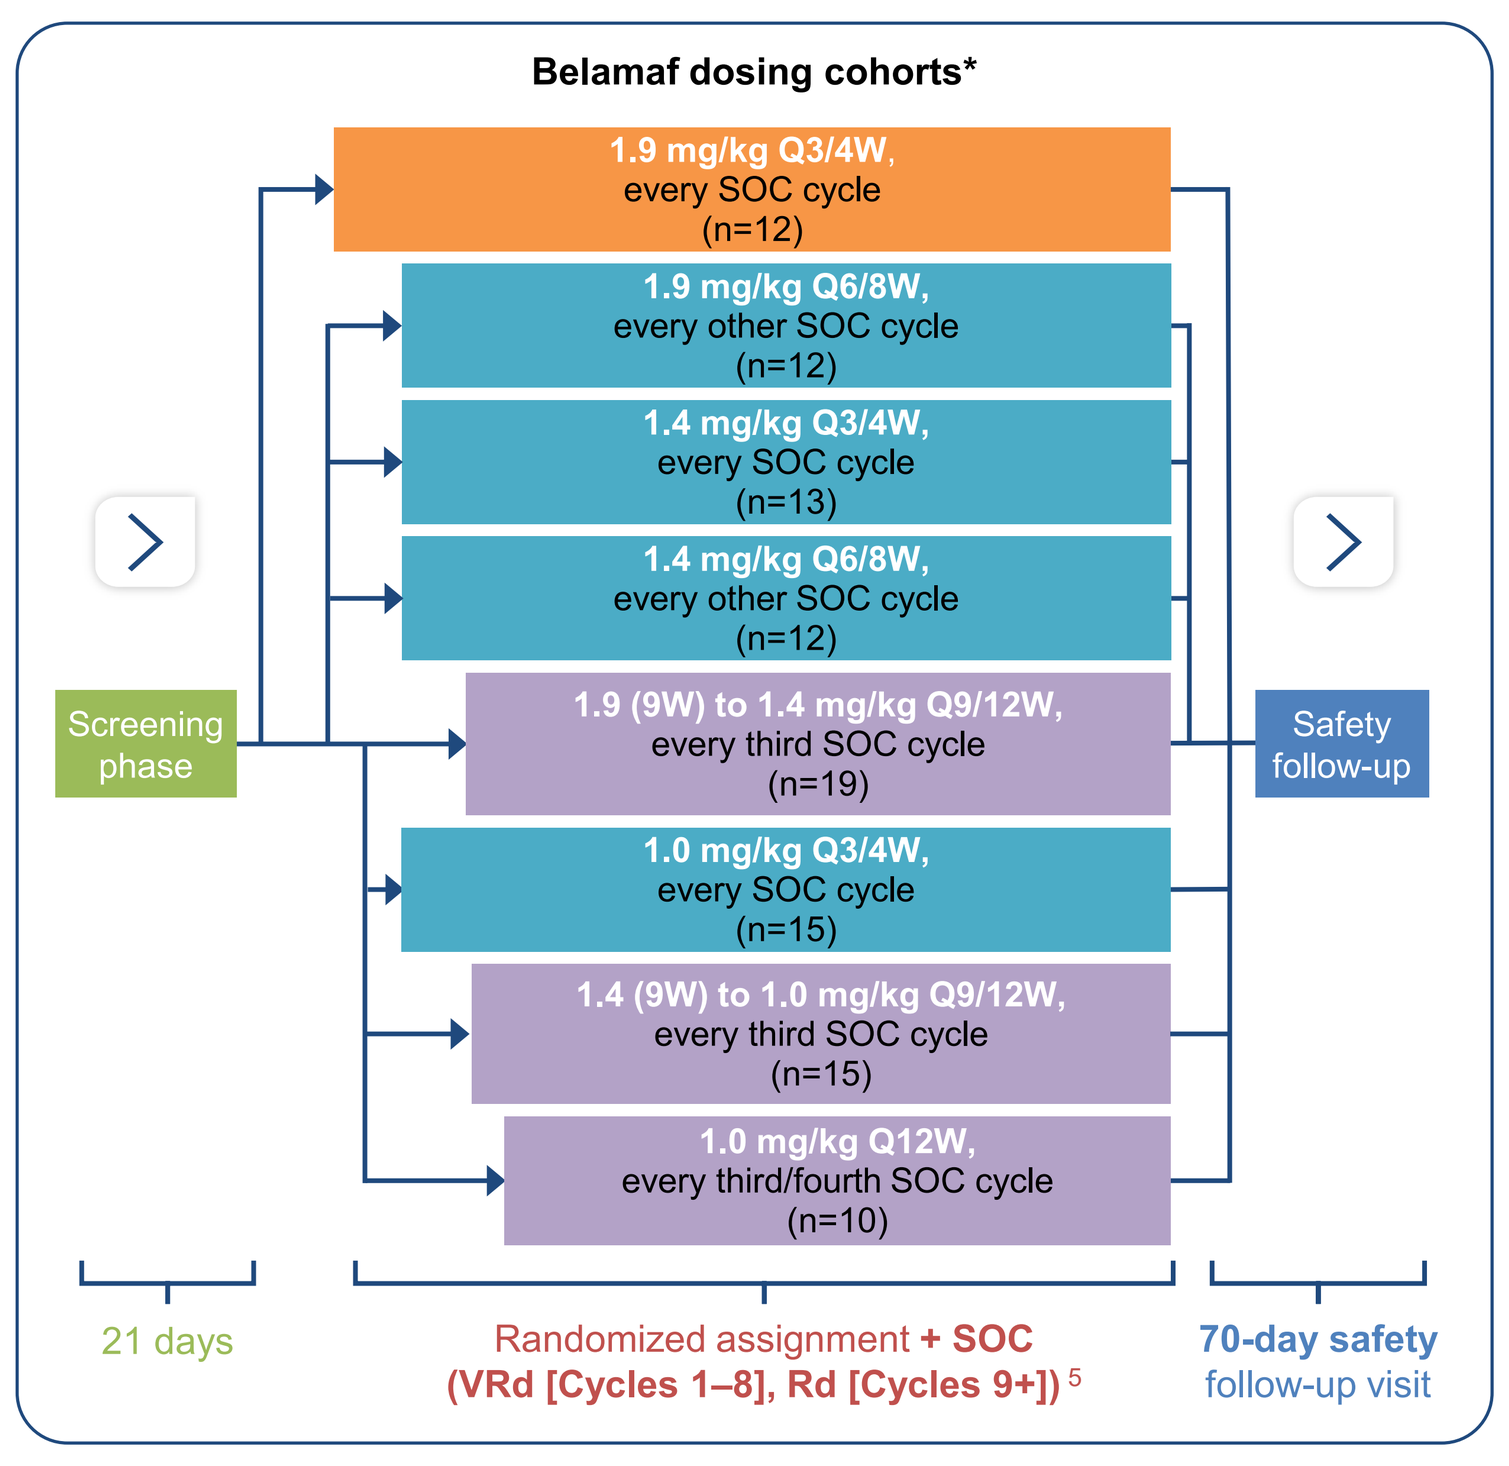
Belamaf dosing cohorts*
1.9 mg/kg Q3/4W, every SOC cycle (n=12)
1.9 mg/kg Q6/8W, every other SOC cycle (n=12)
1.4 mg/kg Q3/4W, every SOC cycle (n=13)
1.4 mg/kg Q6/8W, every other SOC cycle (n=12)
1.9 (9W) to 1.4 mg/kg Q9/12W, every third SOC cycle (n=19)
1.0 mg/kg Q3/4W, every SOC cycle (n=15)
1.4 (9W) to 1.0 mg/kg Q9/12W, every third SOC cycle (n=15)
1.0 mg/kg Q12W, every third/fourth SOC cycle (n=10)
Screening phase
Safety follow-up
21 days
Randomized assignment + SOC (VRd [Cycles 1–8], Rd [Cycles 9+])[5]
70-day safety follow-up visit

Primary endpoint:

- Safety (DLTs and AEs)

Secondary endpoints

- Efficacy (ORR, ≥CR, ≥VGPR per IMWG criteria)
- RDI of lenalidomide and bortezomib after 4 cycles
- Cumulative administered belamaf dose after 4 cycles with VRd
- Incidence and titers of ADAs
- PK

Exploratory endpoint

- MRD negativity (assessed in bone marrow aspirate samples using next-generation sequencing [10^{-5}]).

- Cohorts with lower doses and longer schedules were opened to assess the potential to improve tolerability while maintaining efficacy
- All cohorts received belamaf with VRd for Cycles 1–8 (21-day cycle), followed by belamaf with Rd for Cycles 9+ (28-day cycle)[1]
- Ocular events were managed by dose modifications or extending the dosing interval

*Cohorts of the same color opened at the same time. Cohorts with longer rectangles opened earlier.

RESULTS

Table 1: Patient demographics were comparable across cohorts. Between December 18, 2019 and March 4, 2024 (data cut-off), 108 patients were recruited in 8 cohorts containing 10–19 patients/cohort; enrollment continued through the 2020 coronavirus pandemic. Overall, median age was 74 years, 46% were female, 15% had high-risk cytogenetics, and 11% had EMD

Belamaf schedule	1.9 mg/kg Q3/4W	1.9 mg/kg Q6/8W	1.4 mg/kg Q3/4W	1.4 mg/kg Q6/8W	1.9 (9W) to 1.4 mg/kg Q9/12W	1.0 mg/kg Q3/4W	1.4 (9W) to 1.0 mg/kg Q9/12W	1.0 mg/kg Q12W	Total
	n=12	n=12	n=13	n=12	n=19	n=15	n=15	n=10	N=108
Median age, years (range)	72.5 (63–77)	73.0 (69–78)	74.0 (65–88)	73.0 (69–80)	76.5 (59–88)	73.0 (51–85)	74.0 (52–86)	75.0 (67–85)	74.0 (51–88)
≥75 years, n (%)	5 (42)	4 (33)	6 (46)	4 (33)	13 (68)	3 (20)	6 (40)	6 (60)	47 (44)
Female, n (%)	4 (33)	6 (50)	5 (38)	6 (50)	9 (47)	8 (53)	7 (47)	5 (50)	50 (46)
Ethnicity, n (%)									
White	10 (83)	8 (67)	11 (85)	10 (83)	17 (89)	12 (80)	15 (100)	10 (100)	93 (86)
Black	0	0	0	0	1 (5)	1 (7)	0	0	2 (2)
Asian	2 (17)	4 (33)	2 (15)	2 (17)	1 (5)	2 (13)	0	0	13 (12)
ISS stage at screening, n (%)									
I	2 (17)	5 (42)	7 (54)	3 (25)	5 (26)	6 (40)	7 (47)	2 (20)	37 (34)
II	6 (50)	6 (50)	3 (23)	5 (42)	10 (53)	7 (47)	6 (40)	7 (70)	50 (46)
III	3 (25)	1 (8)	2 (15)	4 (33)	2 (11)	1 (7)	2 (13)	1 (10)	16 (15)
Unknown	1 (8)	0	1 (8)	0	2 (11)	1 (7)	0	0	5 (5)
High-risk cytogenetics*, n (%)	4 (33)	2 (17)	3 (23)	1 (8)	3 (16)	2 (13)	1 (7)	0	16 (15)
EMD, n (%)	2 (17)	3 (25)	0	1 (8)	4 (21)	1 (7)	1 (7)	0	12 (11)

*High cytogenetic risk was defined by the presence of at least one high-risk abnormality: t(4;14), t(14;16), or del(17p13).

Table 2: Rates of Grade 3/4 AEs related to belamaf were lowest among cohorts with lower doses and longer dosing intervals. Of the 105 patients who received ≥1 dose of belamaf, all had an AE, with 33% having a Grade 3/4 AE considered by the investigator to be related to belamaf. The most frequent non-ocular Grade 3+ AEs were thrombocytopenia (30%), neutropenia (26%), and COVID-19 pneumonia (14%), similar to previous studies.[3,4,9,10] Fewer Grade 3/4 AEs were reported during the maintenance phase (50%) than the induction phase (87%)

Belamaf schedule	1.9 mg/kg Q3/4W	1.9 mg/kg Q6/8W	1.4 mg/kg Q3/4W	1.4 mg/kg Q6/8W	1.9 (9W) to 1.4 mg/kg Q9/12W	1.0 mg/kg Q3/4W	1.4 (9W) to 1.0 mg/kg Q9/12W	1.0 mg/kg Q12W	Total
n	12	12	13	12	17	14	15	10	N=105
Median follow-up, months (range)	37.6 (7–50)	32.3 (6–38)	20.2 (1–37)	32.4 (5–37)	17.1 (1–23)	31.0 (0–38)	18.2 (2–22)	7.8 (5–10)	–
Any AE, n (%)	12 (100)	12 (100)	13 (100)	12 (100)	17 (100)	14 (100)	15 (100)	10 (100)	105 (100)
Overall Grade 3/4	12 (100)	12 (100)	12 (92)	12 (100)	15 (88)	13 (93)	15 (100)	7 (70)	98 (93)
Infections/infestations	5 (42)	7 (58)	3 (23)	3 (25)	5 (29)	4 (29)	5 (33)	3 (30)	35 (33)
Grade 3/4 AEs related to belamaf, n (%)	8 (67)	3 (25)	5 (38)	4 (33)	6 (35)	4 (29)	4 (27)	1 (10)	35 (33)
SAE	11 (92)	10 (83)	6 (46)	10 (83)	9 (53)	8 (57)	10 (67)	6 (60)	70 (67)
Fatal SAE related to study treatment	0	0	1 (8)*	1 (8)†	0	0	0	0	2 (2)
Leading to discontinuation of any study treatment	6 (50)	5 (42)	7 (54)	6 (50)	5 (29)	5 (36)	10 (67)	2 (20)	46 (44)
Reason for discontinuation of study treatments, n (%)									
Adverse event	5 (42)	3 (25)	7 (54)	6 (50)	4 (21)	3 (20)	7 (47)	0	35 (32)
Physician decision	2 (17)	1 (8)	2 (15)	2 (17)	1 (5)	2 (13)	0	0	10 (9)
Progressive disease	0	0	1 (8)	1 (8)	1 (5)	0	1 (7)	0	4 (4)
Reached protocol-defined stopping criteria	1 (8)	0	0	0	0	0	0	0	1 (<1)
Withdrawal by patient	5 (42)	2 (17)	2 (15)	0	0	4 (27)	2 (13)	0	15 (14)

*Pneumonia, considered related to belamaf, dexamethasone, and lenalidomide; †Pulmonary sepsis, COVID-19 pneumonia, and *Haemophilius influenzae* pneumonia were recorded as fatal SAEs for the same patient, and all were considered related to lenalidomide.

Table 3: Cohorts with lower doses and longer dosing intervals generally had lower rates of Grade 3/4 KVA events. Maximum Grade 1 KVA events were reported in 11% of patients, maximum Grade 2 KVA events in 19%, and maximum Grade 3/4 KVA events in 55% of patients. Of 2142 KVA assessments, only 15% were Grade 3/4 events

Belamaf schedule	1.9 mg/kg Q3/4W	1.9 mg/kg Q6/8W	1.4 mg/kg Q3/4W	1.4 mg/kg Q6/8W	1.9 (9W) to 1.4 mg/kg Q9/12W	1.0 mg/kg Q3/4W	1.4 (9W) to 1.0 mg/kg Q9/12W	1.0 mg/kg Q12W	Total
n	12	12	13	12	17	14	15	10	N=105
Median follow-up, months (range)	37.6 (7–50)	32.3 (6–38)	20.2 (1–37)	32.4 (5–37)	17.1 (1–23)	31.0 (0–38)	18.2 (2–22)	7.8 (5–10)	–
Grade 3/4 KVA events, n (%)	10 (83)	11 (92)	11 (85)	9 (75)	5 (29)	9 (64)	1 (7)	2 (20)	58 (55)
Grade 4 KVA events, n (%)	4 (33)	0	4 (31)	2 (17)	0	2 (14)	0	0	12 (11)
Total Grade 3/4 KVA events, no. of events (% of all assessments)	97 (26)	36 (10)	42 (14)	50 (18)	14 (6)	73 (22)	10 (4)	2 (3)	324 (15)
Total Grade 4 KVA events, no. of events (% of all assessments)	13 (3)	0	6 (2)	3 (1)	0	5 (2)	0	0	27 (1)
Discontinuation due to Grade ≥3 KVA events, n (%)	1 (8)	0	2 (15)	0	0	2 (14)	0	0	5 (5)

Figure 2: Dose and schedule affected the time to, and resolution of, BCVA decreases. Extending the dosing interval between the 1.9 mg/kg or 1.4 mg/kg doses from Q3/4W to Q6/8W was associated with longer time to BCVA decrease to 20/50 or worse* Resolution of BCVA decreases was generally faster in cohorts with lower initial doses of belamaf

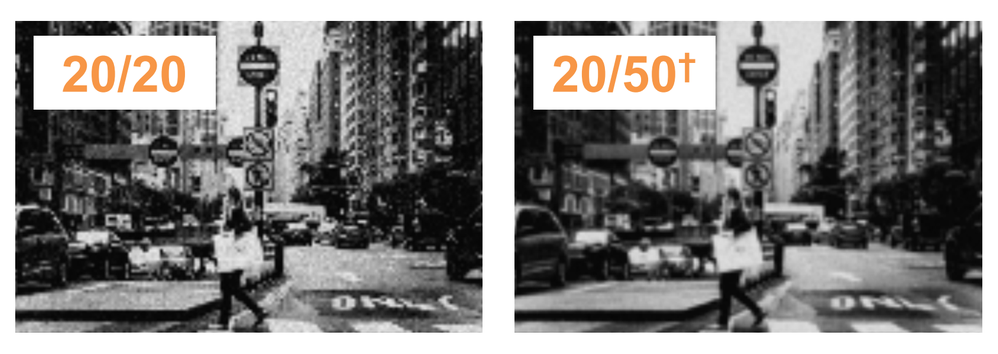
20/20
20/50†

First occurrence of decrease in BCVA score from baseline (20/25 or better) to 20/50 or worse

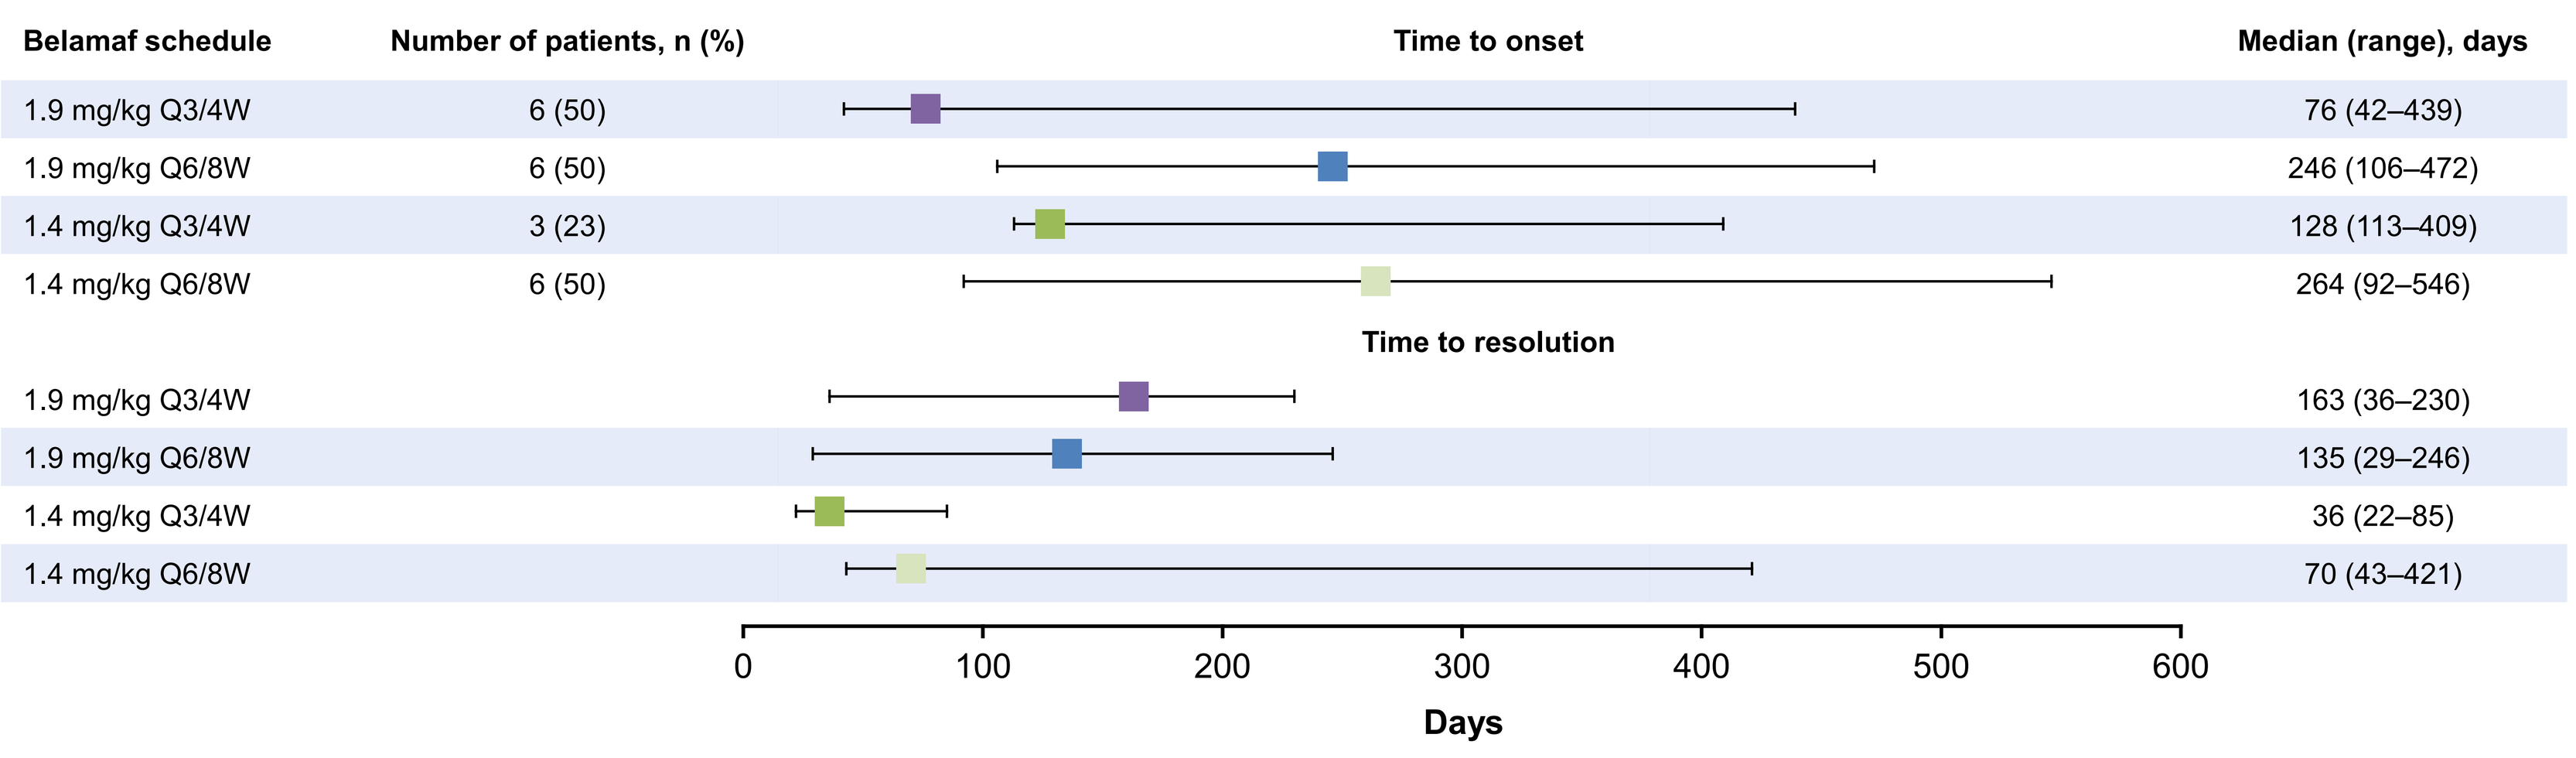
Belamaf schedule
Number of patients, n (%)
Time to onset
Median (range), days
1.9 mg/kg Q3/4W 6 (50) 76 (42–439)
1.9 mg/kg Q6/8W 6 (50) 246 (106–472)
1.4 mg/kg Q3/4W 3 (23) 128 (113–409)
1.4 mg/kg Q6/8W 6 (50) 264 (92–546)
Time to resolution
1.9 mg/kg Q3/4W 163 (36–230)
1.9 mg/kg Q6/8W 135 (29–246)
1.4 mg/kg Q3/4W 36 (22–85)
1.4 mg/kg Q6/8W 70 (43–421)
0
100
200
300
400
500
600
Days

*In the 4 cohorts shown, 2 patients had a BCVA change from 20/25 or better to 20/200 or worse. These patients both had bilateral cataracts.
†Image adapted from Shi C, et al. bioRxiv. 2018;doi:doi.org/10.1101/328443. Copyright © 2018 the Author.

Figure 3: VGPR+ was 100% in 3 cohorts including those with lower doses and less frequent schedules. ORRs ranged from 71% to 100%. Time to achieve VGPR+ was consistent across the cohorts (median 2.1–3.2 months) and response deepened over time. In the first 4 cohorts, CR+ was 62–92%

ORR*, %	100	100	92	100	71	86	87	100
VGPR+ / CR+, %	92 / 75	100 / 92	92 / 62	100 / 91	59 / 41	79 / 71	73 / 53	100 / 30

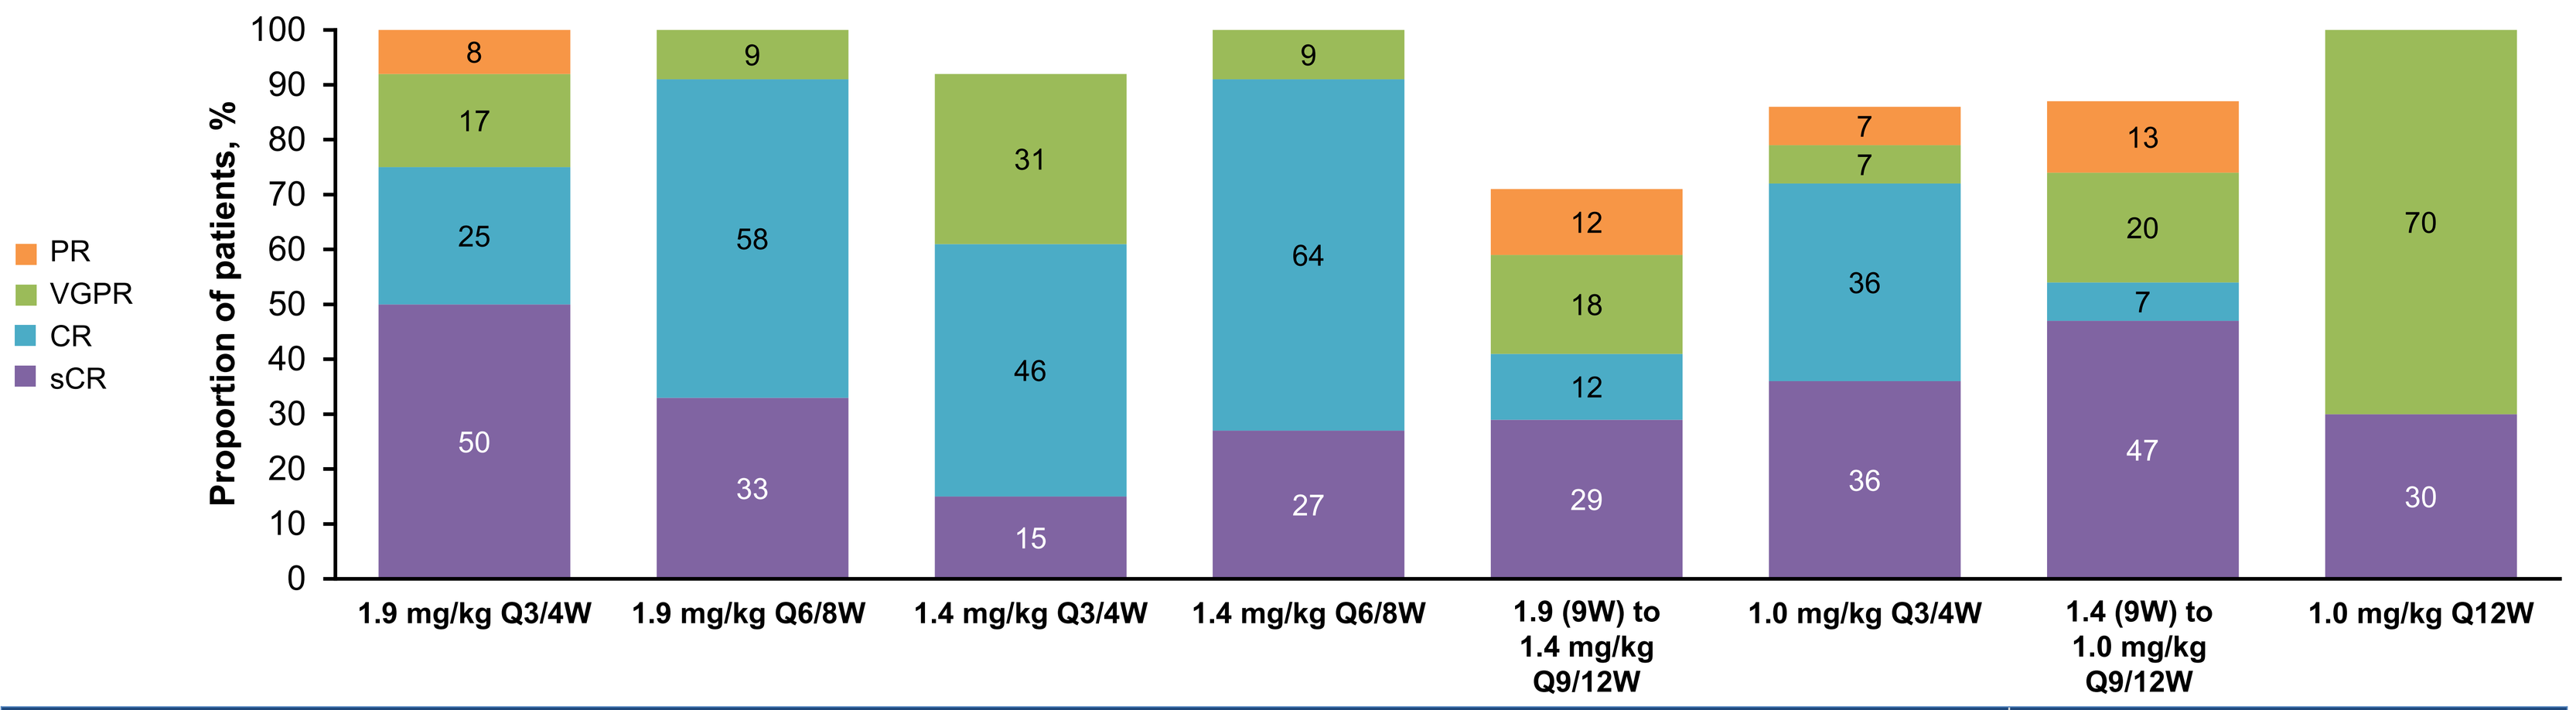
Proportion of patients, %
PR
VGPR
CR
sCR
1.9 mg/kg Q3/4W: 8, 17, 25, 50
1.9 mg/kg Q6/8W: 9, 58, 33
1.4 mg/kg Q3/4W: 31, 46, 15
1.4 mg/kg Q6/8W: 9, 64, 27
1.9 (9W) to 1.4 mg/kg Q9/12W: 12, 18, 12, 29
1.0 mg/kg Q3/4W: 7, 7, 36, 36
1.4 (9W) to 1.0 mg/kg Q9/12W: 13, 20, 7, 47
1.0 mg/kg Q12W: 70, 30

	n=12	n=12	n=13	n=11	n=17	n=14	n=15	n=10
Median follow-up, months (range)	37.6 (7–50)	32.3 (6–38)	20.2 (1–37)	32.4 (5–37)	17.1 (1–23)	31.0 (0–38)	18.2 (2–22)	7.8 (5–10)
Median time to VGPR+, months (range)	2.8 (0.7–4.2)	2.9 (0.8–14.7)	2.2 (0.8–16.8)	2.1 (0.8–5.0)	2.2 (0.7–8.1)	3.0 (1.4–6.8)	3.1 (0.8–4.9)	3.2 (1.3–4.9)
Median time to CR+, months (range)	8.3 (2.1–19.4)	12.0 (4.4–22.9)	6.2 (1.4–15.7)	8.8 (1.4–18.4)	9.5 (3.7–11.1)	8.2 (2.1–19.8)	6.1 (1.4–13.8)	6.5 (2.1–9.1)

Median follow-up varied as cohorts opened at different times; some patients had not been treated for long enough to achieve response

Figure 4: Higher belamaf starting doses were associated with deeper and faster MRD[−] rates*

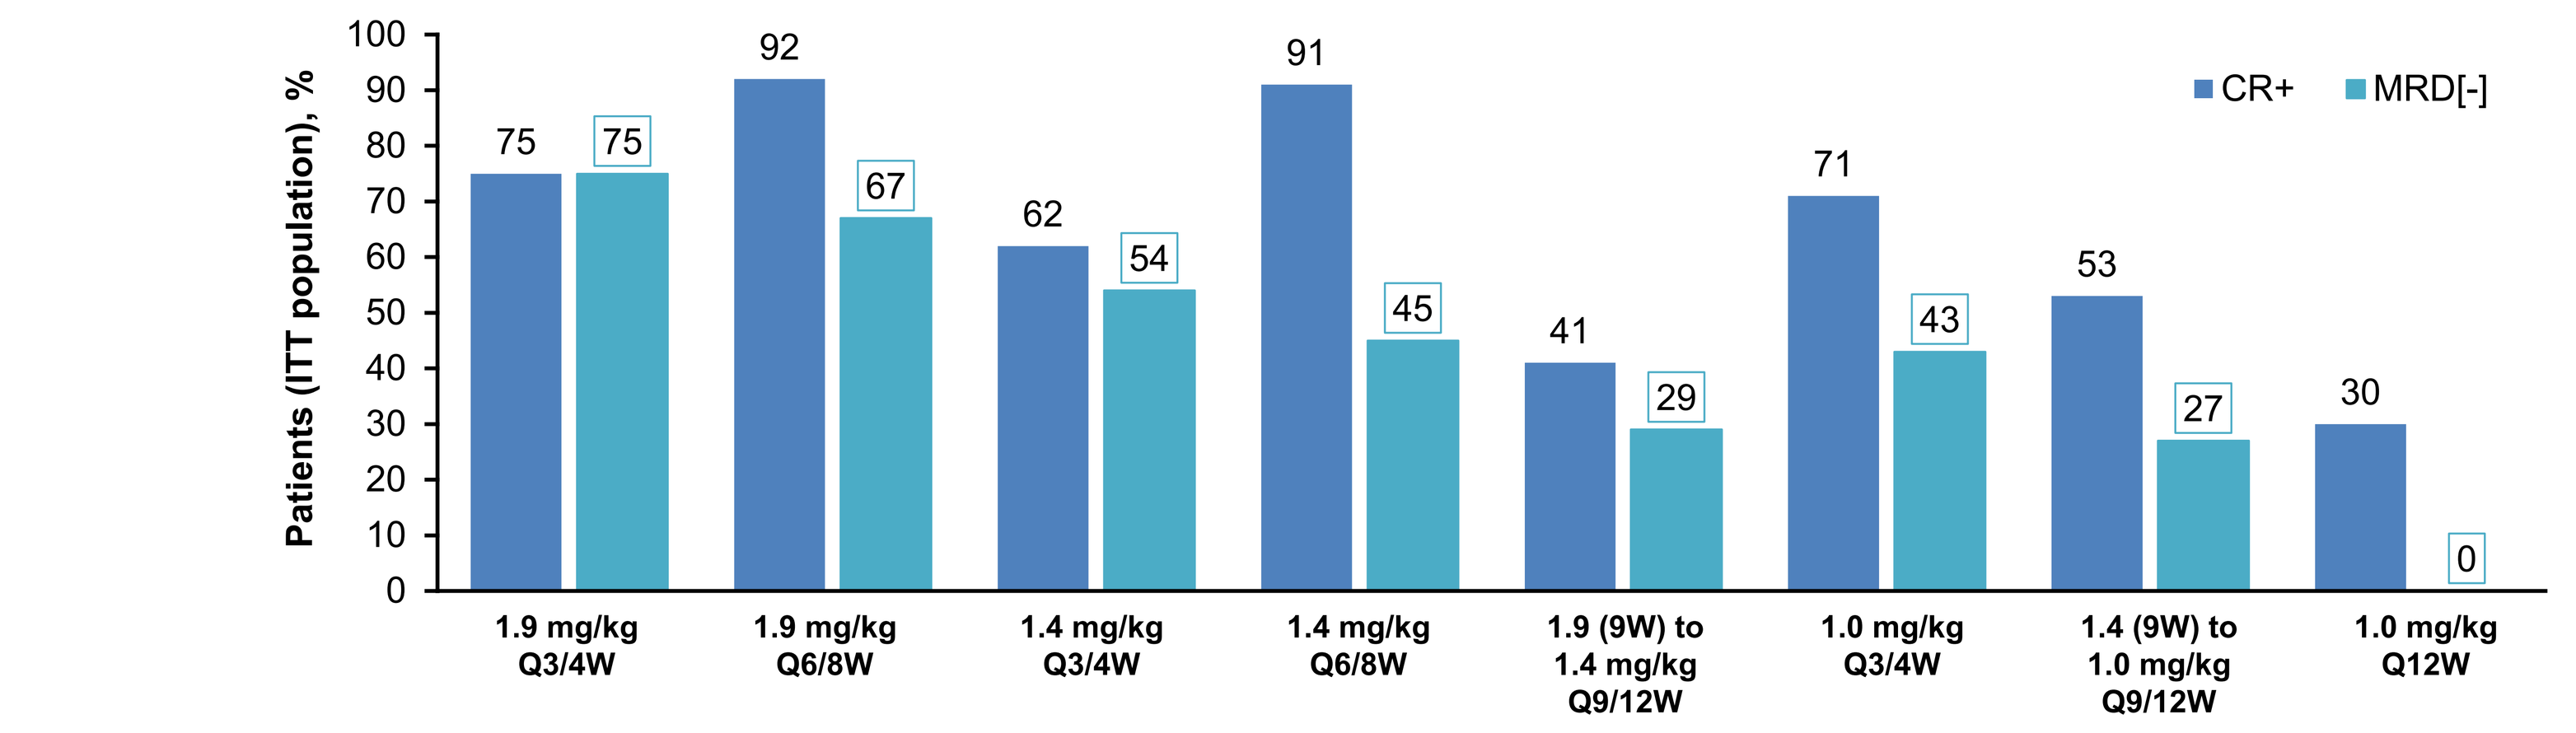
Patients (ITT population), %
CR+
MRD[-]
1.9 mg/kg Q3/4W: 75, 75
1.9 mg/kg Q6/8W: 92, 67
1.4 mg/kg Q3/4W: 62, 54
1.4 mg/kg Q6/8W: 91, 45
1.9 (9W) to 1.4 mg/kg Q9/12W: 41, 29
1.0 mg/kg Q3/4W: 71, 43
1.4 (9W) to 1.0 mg/kg Q9/12W: 53, 27
1.0 mg/kg Q12W: 30, 0

	n=12	n=12	n=13	n=11	n=17	n=14	n=15	n=10
MRD[−], n (% of patients with CR+)	9 (100)	8 (73)	7 (88)	5 (50)	5 (71)	6 (60)	4 (50)	0
Median time to MRD[−], months (range)	8.3 (2.1–17.5)	7.9 (4.2–14.7)	12.2 (2.3–24.9)	14.6 (2.1–23.1)	4.4 (2.2–10.2)	3.8 (2.5–12.3)	5.1 (2.1–9.2)	0
Median follow-up, months	37.6	32.3	20.2	32.4	17	31.0	18.2	7.8

MRD[-] was measured by next-generation sequencing [10^{-5}] in patients achieving CR+, and is shown as proportion of the ITT population.

Figure 5: Higher belamaf starting doses and shorter dosing intervals achieved a higher MRD[−] rate.* MRD[−] rate continues to increase into the Maintenance phase, regardless of dose modifications used to manage KVA Grade 2+ events (30%).† Longer belamaf dosing intervals allowed for a higher mg/kg/cycle to be given

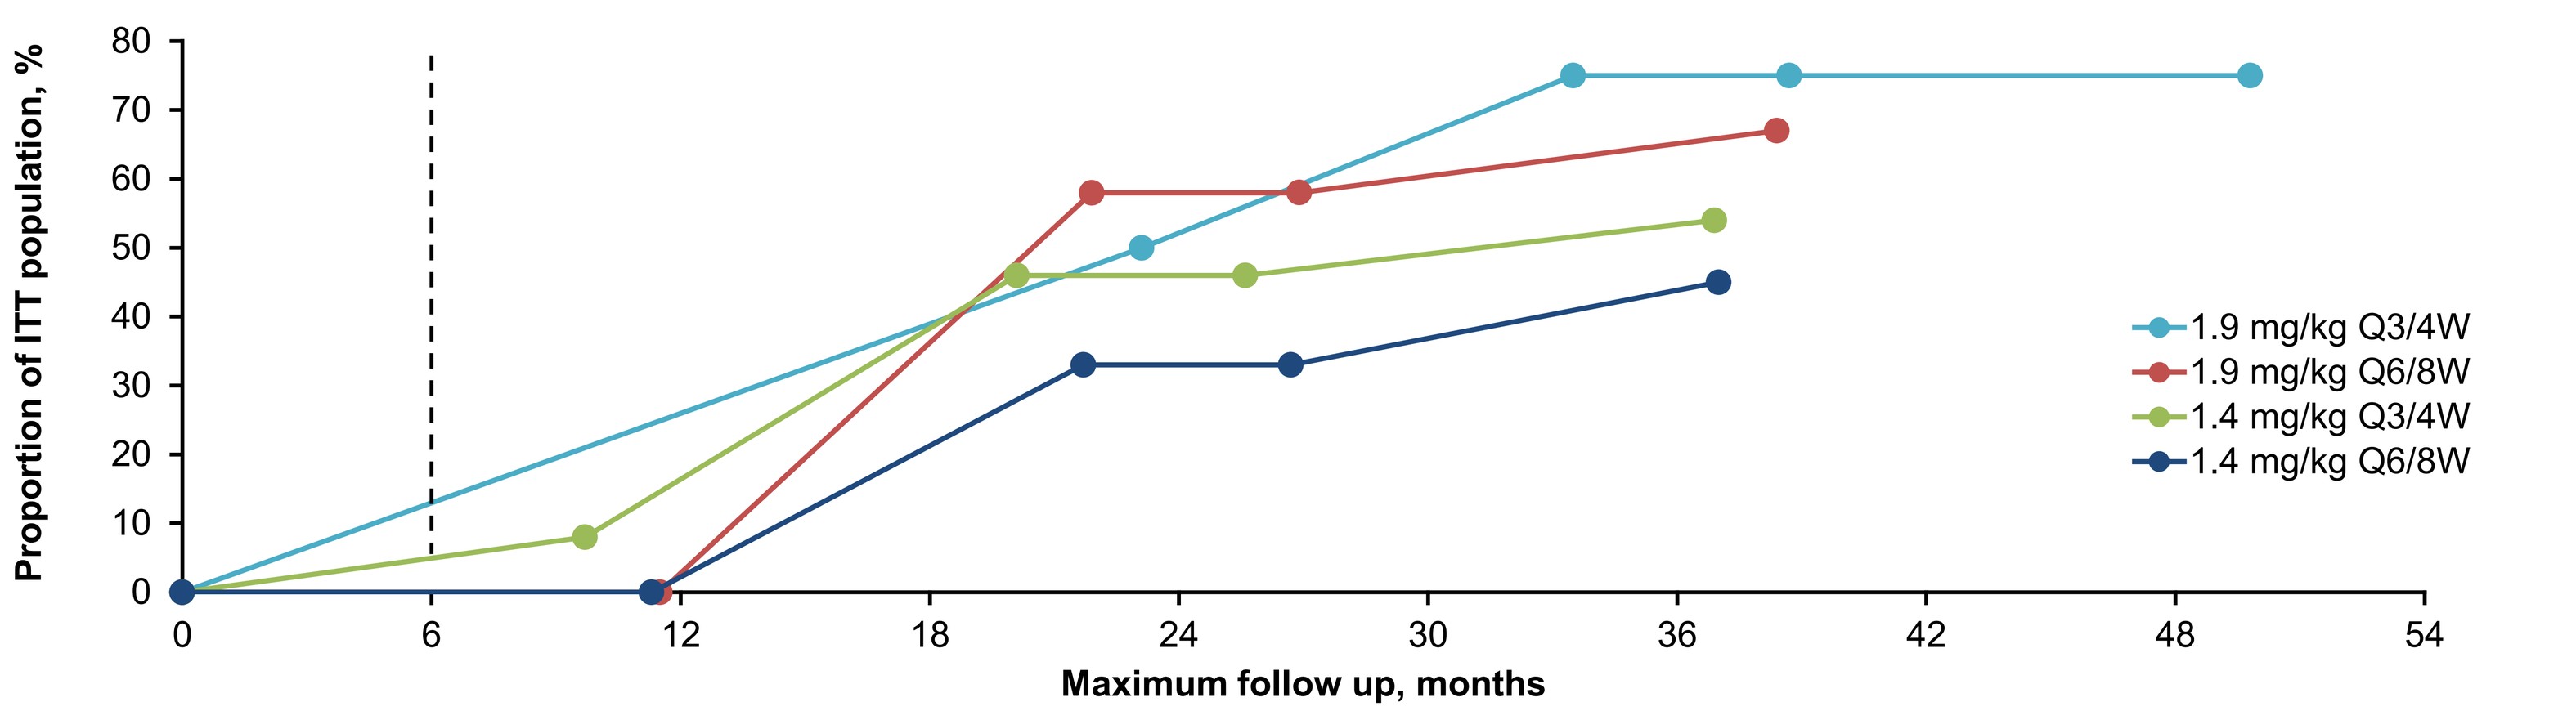
Proportion of ITT population, %
Maximum follow up, months
1.9 mg/kg Q3/4W
1.9 mg/kg Q6/8W
1.4 mg/kg Q3/4W
1.4 mg/kg Q6/8W

Belamaf schedule	End of induction (BVRd) 1.9 mg/kg Q3/4W	1.9 mg/kg Q6/8W	1.4 mg/kg Q3/4W	1.4 mg/kg Q6/8W	Maintenance (BRd) 1.9 mg/kg Q3/4W	1.9 mg/kg Q6/8W	1.4 mg/kg Q3/4W	1.4 mg/kg Q6/8W
Belamaf median RDI, %	59.3	90.4	70.8	91.9	19.6	31.5	27.1	49.1
Belamaf mg/kg/cycle, median	1.13	1.72	0.99	1.29	0.37	0.60	0.38	0.69
Bortezomib median RDI‡, %	53.6	70.2	79.0	77.1	–	–	–	–
Lenalidomide median RDI‡, %	84.2	86.9	91.2	89.0	54.8	53.1	68.7	57.3

*MRD[−] was measured by next-generation sequencing [10^{-5}] in patients achieving CR+; †Belamaf dose Interruptions (2%), reductions (30%), and delays (55%) for Grade 2+ KVA event occurred in 58% of patients overall; ‡Bortezomib and lenalidomide RDI data were analyzed at the previous data cut (March 27, 2023).

CONCLUSIONS

- Across all dosing schedules, belamaf + VRd delivered highly effective tumor responses and MRD negativity in patients with TI NDMM
 - ORR was 71–100% across cohorts with 4 cohorts having 100% ORR. CR+ was 30–92% across cohorts
 - MRD[-] was 0–75% across cohorts: MRD[-] continued to increase into the Maintenance phase
- Higher starting doses of belamaf were generally associated with higher and faster rates of MRD[-] across dosing intervals
 - MRD[-] was 75% in 1.9 mg/kg Q3/4W and 67% in 1.9 mg/kg Q6/8W cohorts
- In cohorts with comparable follow-up durations, longer dosing intervals were associated with increased time to onset of clinically meaningful BCVA changes
- Ocular events were effectively managed with dose modification, including extending the dosing interval or dose reduction, while maintaining patients on treatment across all cohorts. These data are consistent with prior clinical studies of belamaf in the relapsed/refractory MM setting[3,4,9,10]
- A phase 3, randomized, study of belamaf + Rd (BRd) versus daratumumab + Rd (DRd) in patients with TI NDMM (NCT06679101, DREAMM-10) is ongoing[11]

REFERENCES

1. Tai YT, et al. *Blood*. 2014;123:3128–38
2. Tai YT, Anderson KC. *Immunotherapy*. 2015;7:1187
3. Hungria P, et al. *N Engl J Med*. 2024: 391:393–407
4. Dimopoulos AM, et al. *N Engl J Med*. 2024;391:408–21
5. Durie BGM, et al. *Lancet*. 2017;389: 519–27
6. Durie BGM, et al. *Blood Cancer J*. 2020;10:53
7. https://www.clinicaltrials.gov/study/NCT04091126. [accessed October 2024]
8. Usmani S, et al. *J Clin Oncol*. 2023;41(16 suppl):8018
9. Nooka AK, et al. *Cancer*. 2023;129:3746–60
10. Dimopoulos MA, et al. *Lancet Haematol*. 2023;10:e801–12
11. https://clinicaltrials.gov/study/NCT06679101

ABBREVIATIONS

ADA, anti-drug antibodies; ADC, antibody-drug conjugate; ADCC, antibody-dependent cellular cytotoxicity; ADCP, antibody-dependent cellular phagocytosis; AE, adverse event; ASCT, autologous stem cell transplant; ATP, adenosine triphosphate; B, belantamab mafodotin; BCMA, B-cell maturation antigen; belamaf, belantamab mafodotin; BPd, belantamab mafodotin, pomalidomide, dexamethasone; BVCA, best corrected visual acuity; BVd, belantamab mafodotin, bortezomib, dexamethasone; CI, confidence interval; CR+, complete response or better; COVID-19, coronavirus disease 2020; CR, complete response; CTL, cytotoxic T-lymphocyte; DC, dendritic cell; DLT, dose-limiting toxicities; DVd, daratumumab, bortezomib, dexamethasone; ECOG, Eastern Cooperative Oncology Group; EMD, extramedullary disease; HDT, high-dose chemotherapy; HMGB1, high-mobility group box 1; ICD, immunogenic cell death; IMWG, International Myeloma Working Group; ISS, International Staging System; ITT, intention-to-treat; KVA, Keratopathy and Visual Acuity; MM, multiple myeloma; MRD, minimal residual disease; MRD[−], minimal residual disease negativity; mcMMAF, microtubule inhibitor maleimidocaproyl monomethyl auristatin-F; NCT, national clinical trial; ND, newly diagnosed; NDMM, newly diagnosed multiple myeloma; ORR, overall response rate; PFS, progression-free survival; PK, pharmacokinetics; PR, partial response; PVd, pomalidomide, bortezomib, dexamethasone; QxW, every x weeks; Rd, lenalidomide and dexamethasone; RDI, relative dose intensity; SAE, serious adverse event; sCR, stringent complete response; SCT, stem cell transplant; SMM, smouldering MM; SOC, standard-of-care; TI, transplant-ineligible; VGPR, very good partial response; VGPR+, very good partial response or better; VRd, bortezomib, lenalidomide, and dexamethasone.

ACKNOWLEDGEMENTS

Exposure-response analyses for DREAMM-9 were presented in a poster presentation at ASH 2024 (Carreño F, et al. Abstract 1974). Study funding: GSK (209664, NCT04091126). Drug linker technology licensed from Seagen Inc.; monoclonal antibody produced using POTELLIGENT Technology licensed from BioWa. Medical writing support was provided by Elisabeth Walsby, PhD, CMPP, of Fishawack Indicia, a part of Avalere Health and was funded by GSK. The contents of this poster were previously presented at the 66th American Society of Hematology Annual Meeting and Exposition, December 7–10, 2024 and this poster is presented here with permission and on behalf of the original authors.

DISCLOSURES

SZU has received consultancy fees from AbbVie, Amgen, Bristol-Myers Squibb, Bristol-Myers Squibb – Celgene, EdoPharma, Genentech, Gilead, GSK, Gracell Therapeutics, Johnson & Johnson – Janssen, Oncopeptides, Sanofi, Seagen, SecuraBio, SkylineDX, Takeda, and TeneoBio; and has received research funding from AbbVie, Amgen, Array Biopharma, Bristol-Myers Squibb, Bristol-Myers Squibb – Celgene, GSK, Johnson & Johnson – Janssen, Merck, Pharmacyclics, Sanofi, Seagen, SkylineDX, and Takeda. **MM** has received consultancy fees from GSK, AbbVie, Johnson & Johnson – Janssen; has received honoraria directly from GSK, AbbVie, Johnson & Johnson – Janssen, Amgen, Novartis, Bristol-Myers Squibb – Celgene, Bristol-Myers Squibb, MSD, Sanofi, and Eli Lilly. **MG** has served on advisory boards for Amgen, Bristol-Myers Squibb – Celgene, CTI, Johnson & Johnson – Janssen, Sanofi, and Stemline, and has received speaker fees from Amgen, AOP Pharma, Johnson & Johnson – Janssen, Novartis, and Pfizer. **IS** has received honoraria from Johnson & Johnson – Janssen, Bristol-Myers Squibb – Celgene, Pfizer, Sanofi, Gilead/Kite, Vertex, and GSK. **AOA** has no conflicts of interest to disclose. **YK** has received consultancy fees from GSK, Johnson & Johnson – Janssen, Novartis, and Takeda; holds ownership interests in NOBO Medicine (not publicly traded) and Curocell (not publicly traded); and has received honoraria from Celltrion. **AO** has received consultancy fees from Johnson & Johnson – Janssen, Bristol-Myers Squibb, Pfizer, Sanofi, GSK, and Oncopeptides; and is on the board of directors, speaker's bureau, or advisory committee for Johnson & Johnson – Janssen, Pfizer, Sanofi, and GSK. **HQ** has received consultancy fees from GSK, Karyopharm, Bristol-Myers Squibb, AbbVie, Johnson & Johnson – Janssen, Roche, and Pfizer; and has research funding from GSK, Karyopharm, Bristol-Myers Squibb, AbbVie. **KW** has received research funding from AbbVie, Amgen, Bristol-Myers Squibb – Celgene, GSK, Johnson & Johnson – Janssen, and Sanofi (to institution); has received honoraria from AbbVie, Amgen, Adaptive Biotech, AstraZeneca, Beigene, Bristol-Myers Squibb, Bristol-Myers Squibb – Celgene, Johnson & Johnson – Janssen, GSK, Karyopharm, Menarini, Novartis, Oncopeptides, Pfizer, Roche, Sanofi, Stemline, and Takeda; and is on the board of directors, speaker's bureau, or advisory committee for AbbVie, Amgen, Adaptive Biotech, Beigene, Bristol-Myers Squibb, Bristol-Myers Squibb – Celgene, Johnson & Johnson – Janssen, GSK, Karyopharm, Menarini, Novartis, Oncopeptides, Pfizer, Roche, Sanofi, and Takeda. **AAA** has no conflicts of interest to disclose. EMO has received consultancy fees from AbbVie, AstraZeneca, Bristol-Myers Squibb, GSK, Johnson & Johnson – Janssen, Menarini, Oncopeptides, Pfizer, Sanofi, and Takeda; had received research funding from GSK and Oncopeptides; has received Honoria from AbbVie, Amgen, Bristol-Myers Squibb, GSK, Johnson & Johnson – Janssen, Menarini, Oncopeptides, Pfizer, Regeneron, Sanofi, and Takeda; and is a member of the speaker's bureau for Johnson & Johnson – Janssen. **WJ** has served on advisory boards and received speaker fees from Johnson & Johnson – Janssen, Pfizer, and Beigene. **CKM** has no conflicts of interest to disclose. **KR** has received grant funding (to institution) from Sanofi, GSK, and Bristol-Myers Squibb – Celgene; has received honoraria from Johnson & Johnson – Janssen, Adaptive Biotech, Amgen, Takeda, AbbVie, Oncopeptides, Bristol-Myers Squibb – Celgene, Pfizer, Recordati, and GSK; serves on advisory boards for Johnson & Johnson – Janssen, Adaptive Biotech, Amgen, Recordati, Takeda, AbbVie, Oncopeptides, Bristol-Myers Squibb – Celgene, Pfizer, Menarini Stemline, and GSK; and has received travel support from Bristol-Myers Squibb, Amgen, Takeda, and Recordati. **RGS** has received consultancy fees from Johnson & Johnson – Janssen, Bristol-Myers Squibb, Pfizer, Sanofi, and GSK, and is a member of the speaker's bureau for Johnson & Johnson – Janssen and Sanofi. **PRO** has received consultancy fees from Roche, Bristol-Myers Squibb, Johnson & Johnson – Janssen, Sanofi, GSK, Regeneron, Oncopeptides, AbbVie, and Pfizer; has received research funding (to institution) from Pfizer; and serves on the steering committee for Regeneron, Johnson & Johnson – Janssen, and Bristol-Myers Squibb, and the speaker's bureau for Johnson & Johnson – Janssen. **CB** and **MCK** are employees of and hold financial equities in GSK. **JLE** is an employee of and holds financial equities in GSK; and receives patents and loyalties from MRC. **MH** has received honoraria from GSK, AbbVie, Johnson & Johnson – Janssen, Amgen, Novartis, Bristol-Myers Squibb – Celgene, Bristol-Myers Squibb, MSD, Roche, Pfizer, Sanofi, and Eli Lilly.

CoMy | 15–18 May 2025 | Paris, France
Presenting author: Enrique M Ocio ocioem@unican.es

https://comylive.cme-congresses.com